755

Remoxy™: A New Opioid Drug With Effective Analgesia and Abuse-resistance

Pain Therapeutics, Inc.

L. Gilderman, P.G. Butera, D. Gilmore, L.V. Moran and N. Friedmann

ABSTRACT

Abuse of controlled-release (CR) oxycodone tablets such as Oxycontin® is a serious health concern. Abusers defeat the CR mechanism simply by crushing or dissolving a tablet. Ingesting a fractioned tablet produces a quick, powerful high that can be fatal or lead to opioid addiction. Remoxy™ is a new long-acting oxycodone formulation that is highly resistant to abuse. Its viscous gel-cap cannot be injected and its CR mechanism resists crushing and dissolution in alcohol or acidic beverages. We previously demonstrated that Remoxy does not produce a large spike in serum concentrations of oxycodone compared to Oxycontin® when ingested after common methods of abuse. Here we report the analgesic efficacy of Remoxy. We conducted a randomized, double-blind Phase III study in 209 patients with moderate-to-severe chronic pain due to osteoarthritis of the hip or knee. After a washout period, patients were randomized to Remoxy or placebo if their pain intensity was ≥ 5 on a 0-10 scale. Baseline pain intensity was similar in both groups (7.1). Patients were started on 10 mg BID for one week, and then titrated to 20 mg BID for four weeks. In this study, Remoxy provided superior analgesia to placebo, meeting the study's primary endpoint (p = 0.043). Remoxy patients also reported better function (WOMAC total score, p = 0.045; pain subscale, p = 0.015; stiffness subscale, p = 0.040), better quality of life (SF-12 physical function scale, p<0.001) and better quality of analgesia (60.9% versus 38.4% of patients rated quality of analgesia as excellent, very good or good; p<0.001). Adverse events were limited to common opioid-related side effects. Remoxy provides effective, long-acting analgesia for patients with moderate-to-severe pain while offering abuse-resistance to safeguard against diversion and abuse.

INTRODUCTION

Abuse of controlled-release formulations of oxycodone and other prescription opioids is a large, fast-growing problem in the US. Simply crushing and dissolving these formulations can yield the full 12-hr dose to produce an immediate, large spike in opiate blood levels and a powerful morphine-like high, as well as the potential for respiratory depression and death. In the U.S. in 2002, oxycodone abuse resulted in over 22,000 Emergency Room visits (nearly 20% of ER visits due to abuse of narcotic analgesics), according to the Drug Abuse Warning Network (DAWN), a division of the Substance Abuse and Mental Health Services Administration (SAMHSA). This dramatic increase has coincided with the 1996 introduction and marketing of Oxycontin (Fig. 1, Source: DAWN).

Fig. 1 **Emergency Room Visits Involving Oxycodone Abuse**

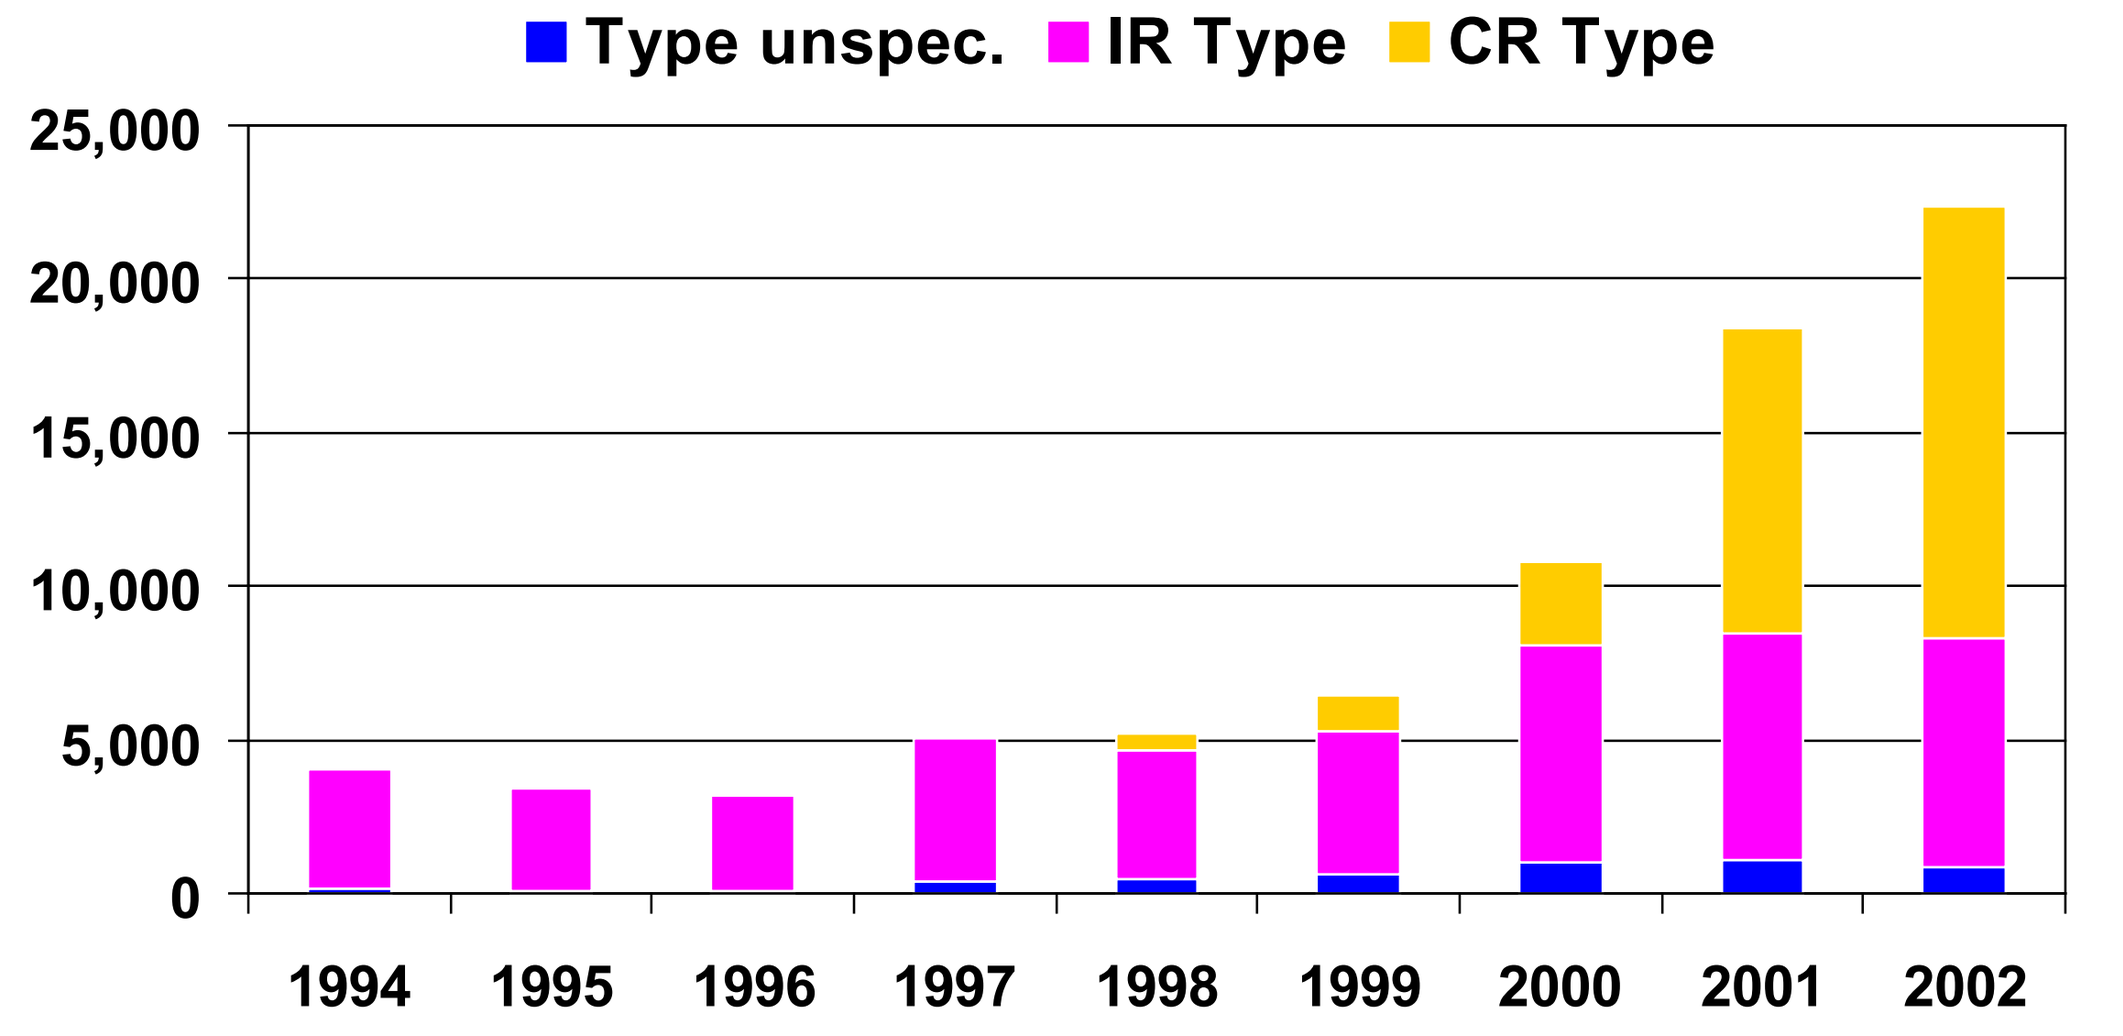

Type unspec.
IR Type
CR Type
25,000
20,000
15,000
10,000
5,000
0
1994
1995
1996
1997
1998
1999
2000
2001
2002

IR = immediate release CR = controlled release

Between 2002 and 2003, the number of people over 12 years old reporting abuse of Oxycontin increased from 1.9 million to 2.8 million, according to the National Survey on Drug Use and Health. Most alarmingly, in a 2005 survey conducted by the National Institute on Drug Abuse (NIDA), 5.5% of 12th-graders reported abusing Oxycontin.

Remoxy is a novel long-acting oxycodone formulation designed to deter oxycodone abuse. In vitro tests show that Remoxy cannot be fragmented by forceful crushing, even after freezing at –80˚C, and that its oxycodone content cannot be extracted by dissolution in alcohol or other common beverages. Ingesting Oxycontin after crushing and dissolving in water or alcohol produced plasma oxycodone levels even slightly higher than ingestion of an equivalent strength immediate release oxycodone tablet. In contrast, oxycodone plasma concentrations following similar treatment of Remoxy remained far below those of the immediate release comparator (Fig. 2)

Fig. 2 **Remoxy vs. Oxycontin**

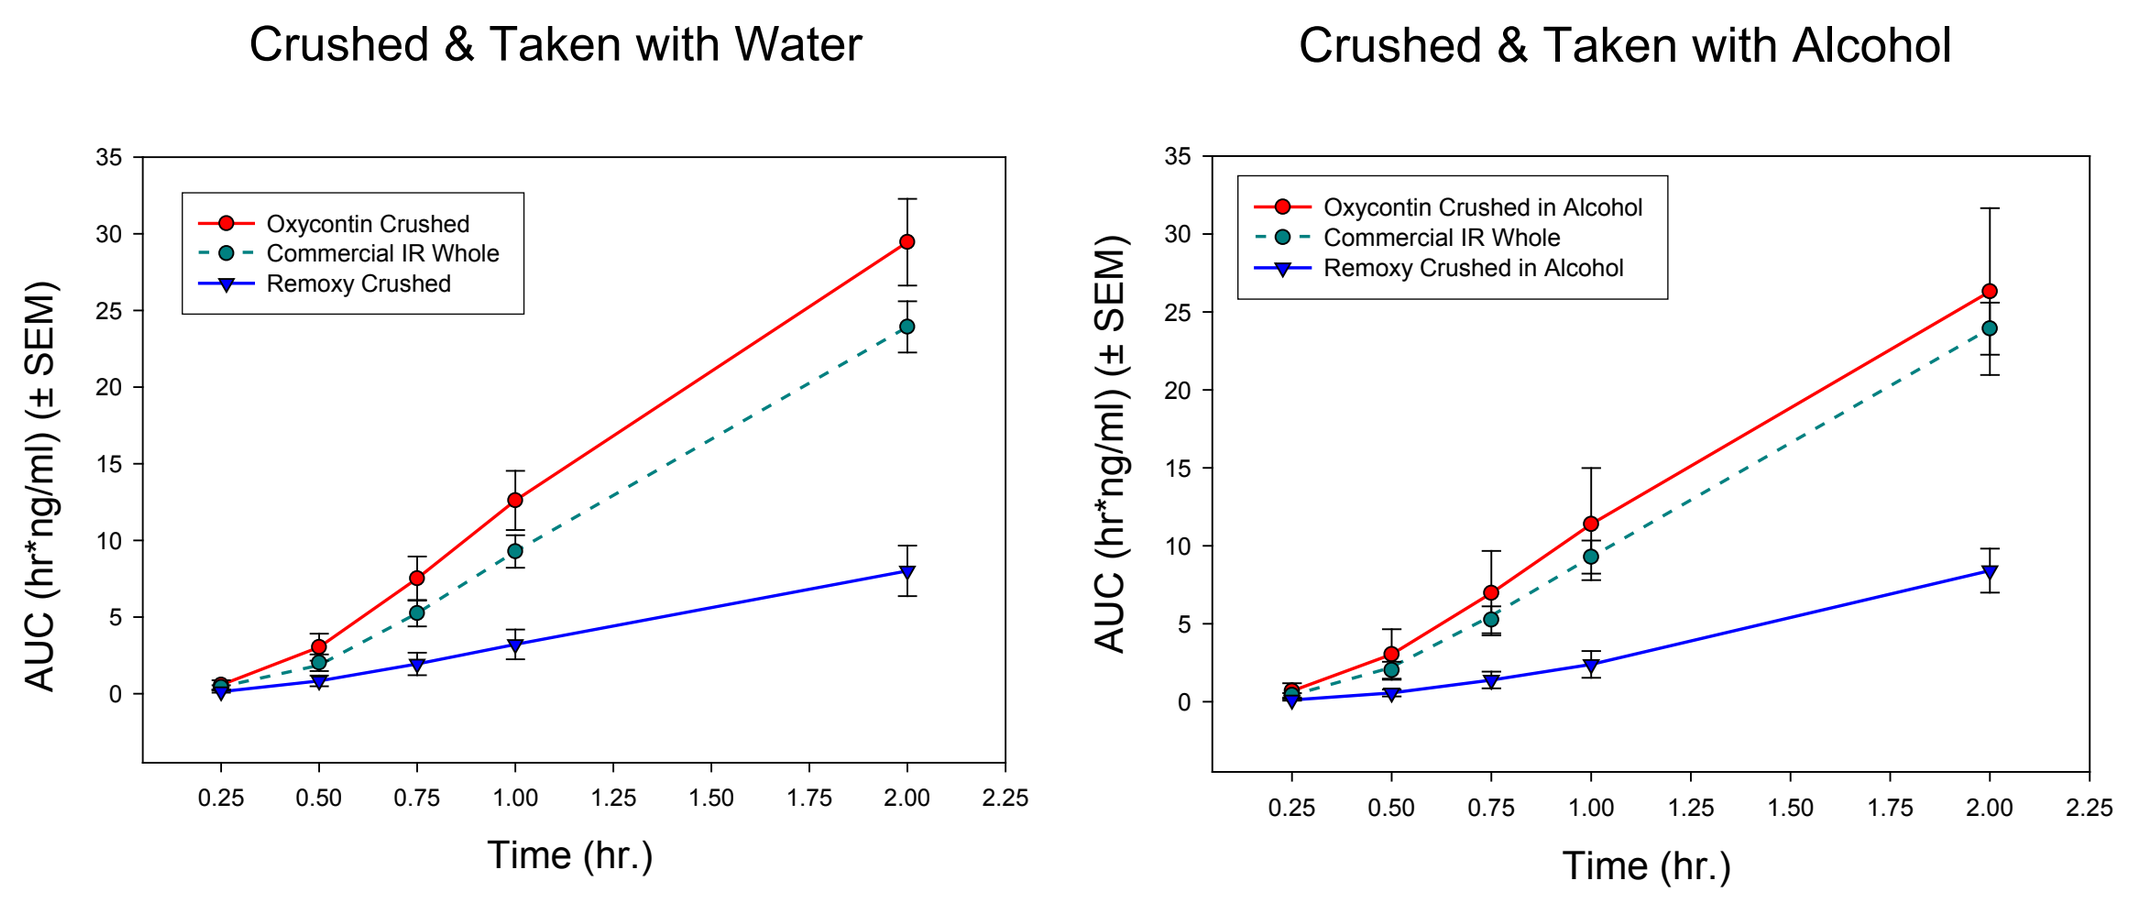

Crushed & Taken with Water
Oxycontin Crushed
Commercial IR Whole
Remoxy Crushed
AUC (hr*ng/ml) (± SEM)
Time (hr.)
Crushed & Taken with Alcohol
Oxycontin Crushed in Alcohol
Commercial IR Whole
Remoxy Crushed in Alcohol
AUC (hr*ng/ml) (± SEM)
Time (hr.)

The current randomized double-blind Phase III clinical trial was designed to assess the analgesic efficacy of Remoxy versus placebo.

METHODS

Patients with moderate-to-severe chronic pain due to osteoarthritis of the hip or knee with a pain intensity (PI) score ≥ 5 on a 0-10 scale at screening entered a 4-to-7 day washout from all analgesics except acetaminophen. Patients with a baseline PI ≥ 5 averaged over the last 2 days of washout were randomized to receive Remoxy or placebo. Patients were started on 10 mg BID for one week and were then titrated up to 20 mg BID for an additional four weeks. Patients recorded their overall PI over the last 24 hours in a daily diary. Adverse events were recorded at weekly clinic visits.

Efficacy Measures

Analgesic efficacy was assessed by percent reduction in PI at the end of treatment. Secondary efficacy measures included the Short Form 12 Question Health Survey (SF-12) and the Western Ontario and MacMaster Universities (WOMAC) Osteoarthritis Index subscales and total score to indicate quality of life, and patient assessment of overall Quality of Analgesia and Global Assessment of Study Medication at the end of treatment.

Statistics

The percent change in PI was analyzed by analysis of covariance (ANCOVA) with treatment, age and sex as effects and baseline PI as the covariate, utilizing the last observation carried forward (LOCF) imputation method for missing values.

Two different sensitivity analyses were used to confirm results using the LOCF imputation method: the first imputed missing PI values as the mean PI among all patients with the same previous value, and the second used the mean PI among patients within the same treatment group with the same previous PI. In addition, the area under the curve (AUC) for the change in PI was performed using a mixed effects model for repeated measures (MMRM). Linear contrasts were constructed to estimate the mean AUC for PI change from baseline vs. time, i.e., linear combinations of the MMRM model parameters were formed corresponding to the appropriate time-weighted sum arising from the trapezoidal rule.

For secondary efficacy analyses, percent change from baseline values of SF-12 and WOMAC were analyzed using ANCOVA models with baseline score as a covariate. Global Assessment and Quality of Analgesia ratings were analyzed using the Cochran-Mantel-Haenszel row mean scores (CMH-RMS) test using equally spaced scores.

RESULTS

Patients

Demographic characteristics and baseline PI (7.1) were similar for both groups. Out of the 209 patients randomized to treatment, 62 (29.7%) discontinued early. Adverse events, mostly opioid-related, was the most common reason for discontinuation in the Remoxy treatment group (24.8% of patients), while inadequate pain relief was the most common reason for discontinuation in the placebo treatment group (18.3% of patients). A study flow diagram is depicted below (Fig. 3).

Fig. 3 **Study Flow Diagram**

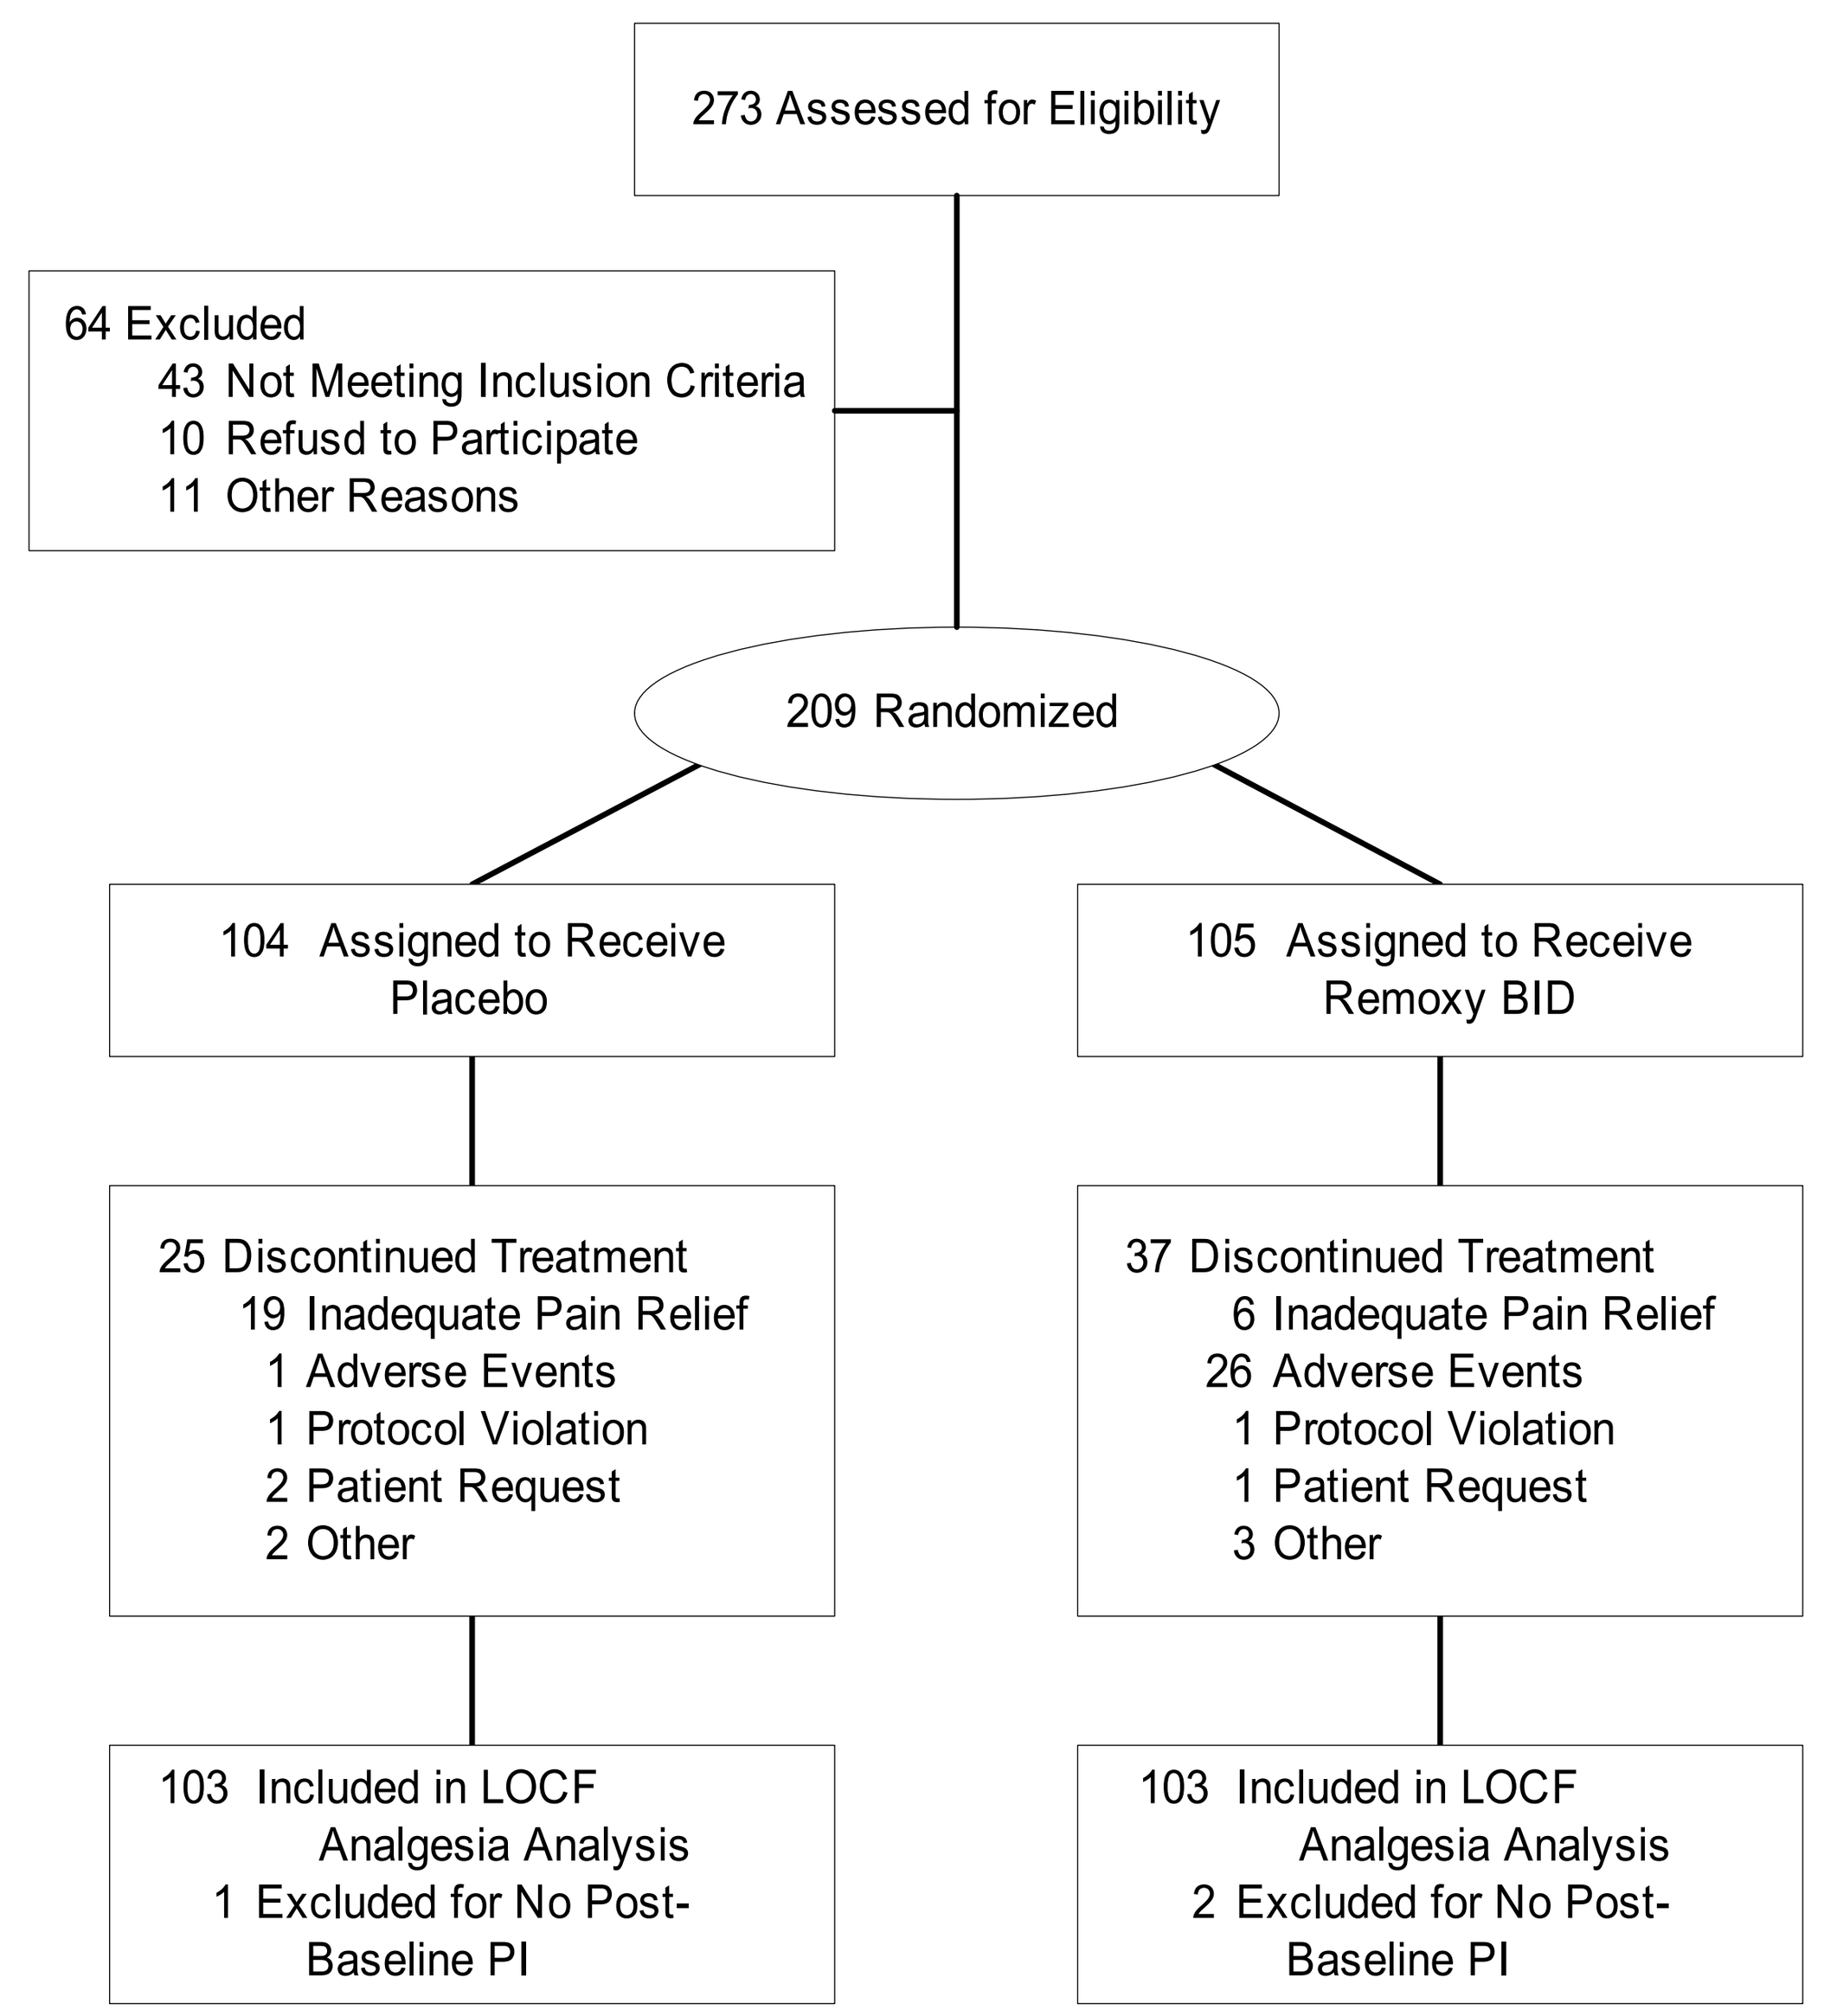

273 Assessed for Eligibility
64 Excluded
43 Not Meeting Inclusion Criteria
10 Refusd to Participate
11 Other Reasons
209 Randomized
104 Assigned to Receive Placebo
105 Assigned to Receive Remoxy BID
25 Discontinued Treatment
19 Inadequate Pain Relief
1 Adverse Events
1 Protocol Violation
2 Patient Request
2 Other
37 Discontinued Treatment
6 Inadequate Pain Relief
26 Adverse Events
1 Protocol Violation
1 Patient Request
3 Other
103 Included in LOCF Analgesia Analysis
1 Excluded for No Post-Baseline PI
103 Included in LOCF Analgesia Analysis
2 Excluded for No Post-Baseline PI

Analgesia

Remoxy provided superior analgesia to placebo as measured by the percent change in PI from baseline at the end of the treatment period (p=0.043; Fig. 4). Sensitivity analyses also revealed significantly greater reductions in pain intensity in the Remoxy treatment group for the average across treatments imputation (p=0.032) and the average within treatment imputation (p=0.034), validating the results of the primary analysis. The AUC for the change in PI from baseline was also significantly greater for Remoxy versus placebo (p=0.013).

Fig. 4 **Percent Reduction in Pain Intensity**

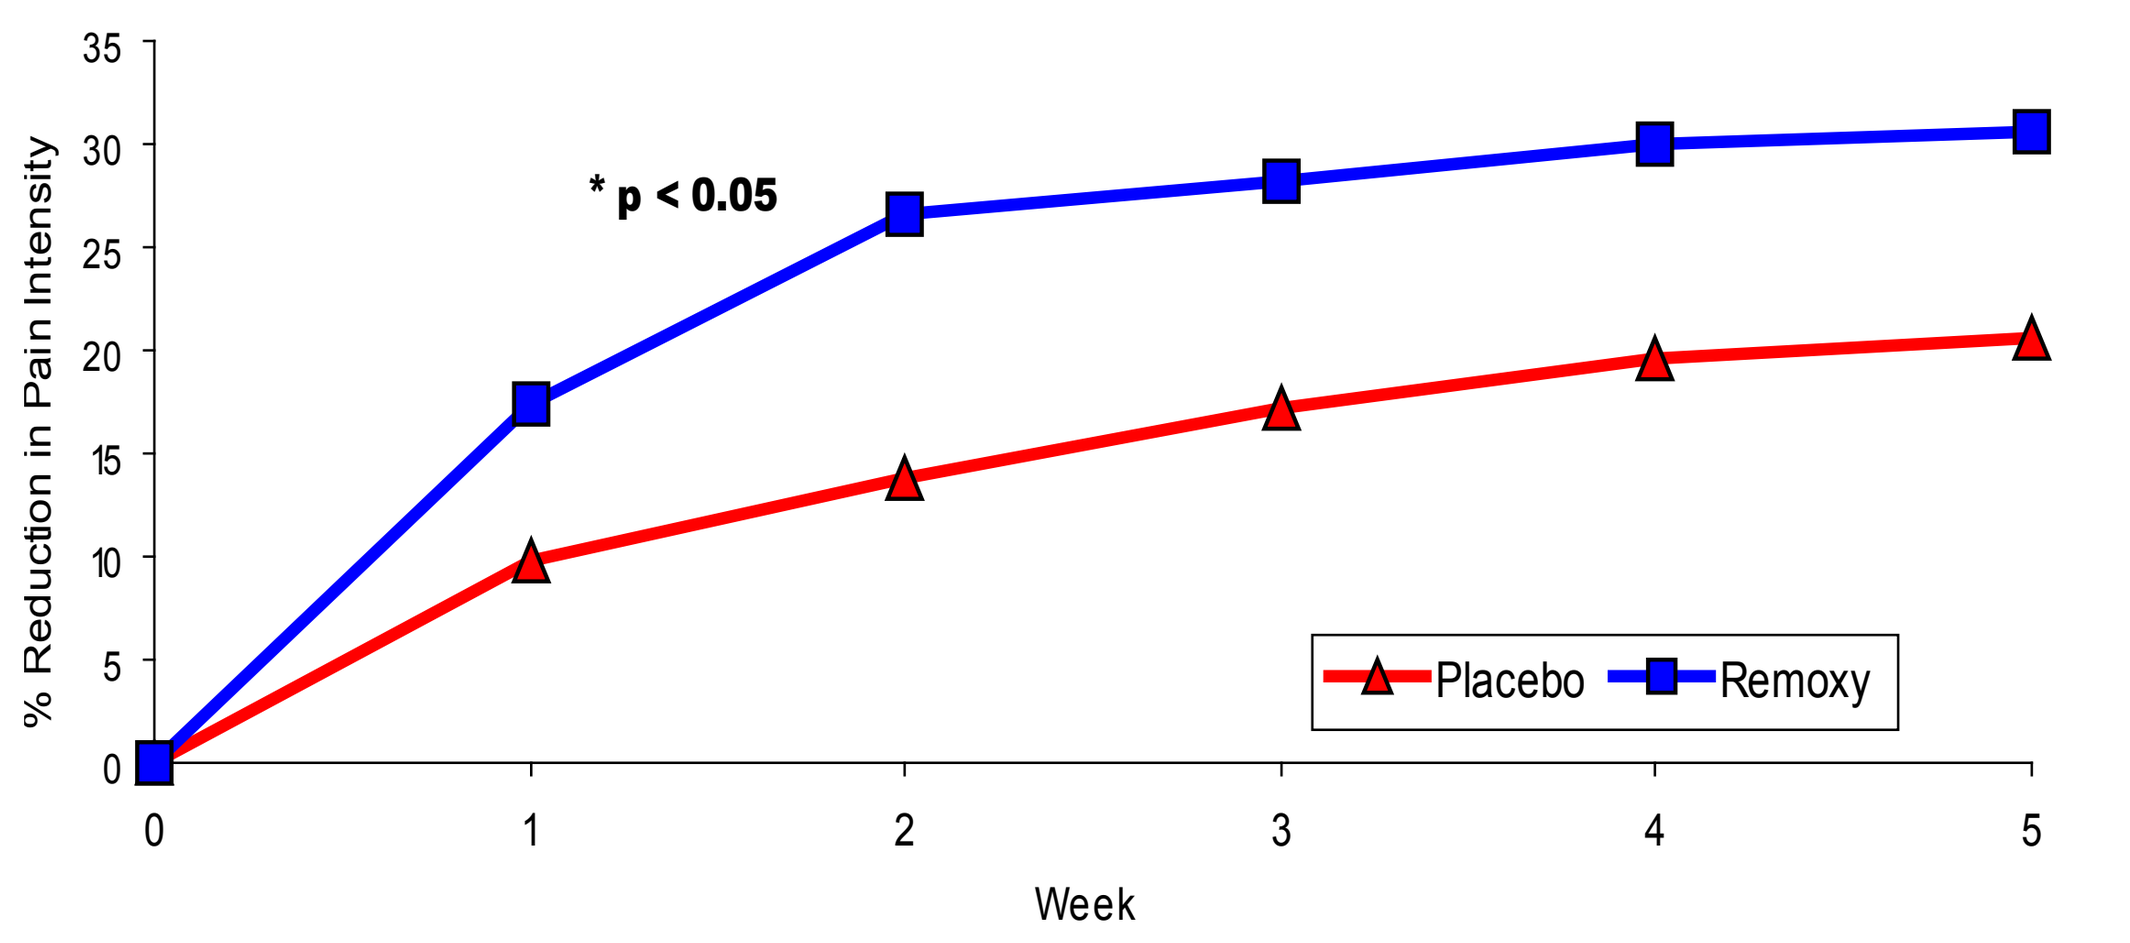

% Reduction in Pain Intensity
* p < 0.05
Placebo
Remoxy
Week
0
1
2
3
4
5

In addition, in patients' assessment of Quality of Analgesia, 60.9% of patients taking Remoxy and 38.4% of placebo patients rated the overall quality of analgesia as excellent, very good, or good (Fig. 5; p<0.001). A trend towards better global assessment of study medication was noted in the Remoxy group at the end of treatment (p=0.097).

Fig. 5 **Quality of Analgesia**

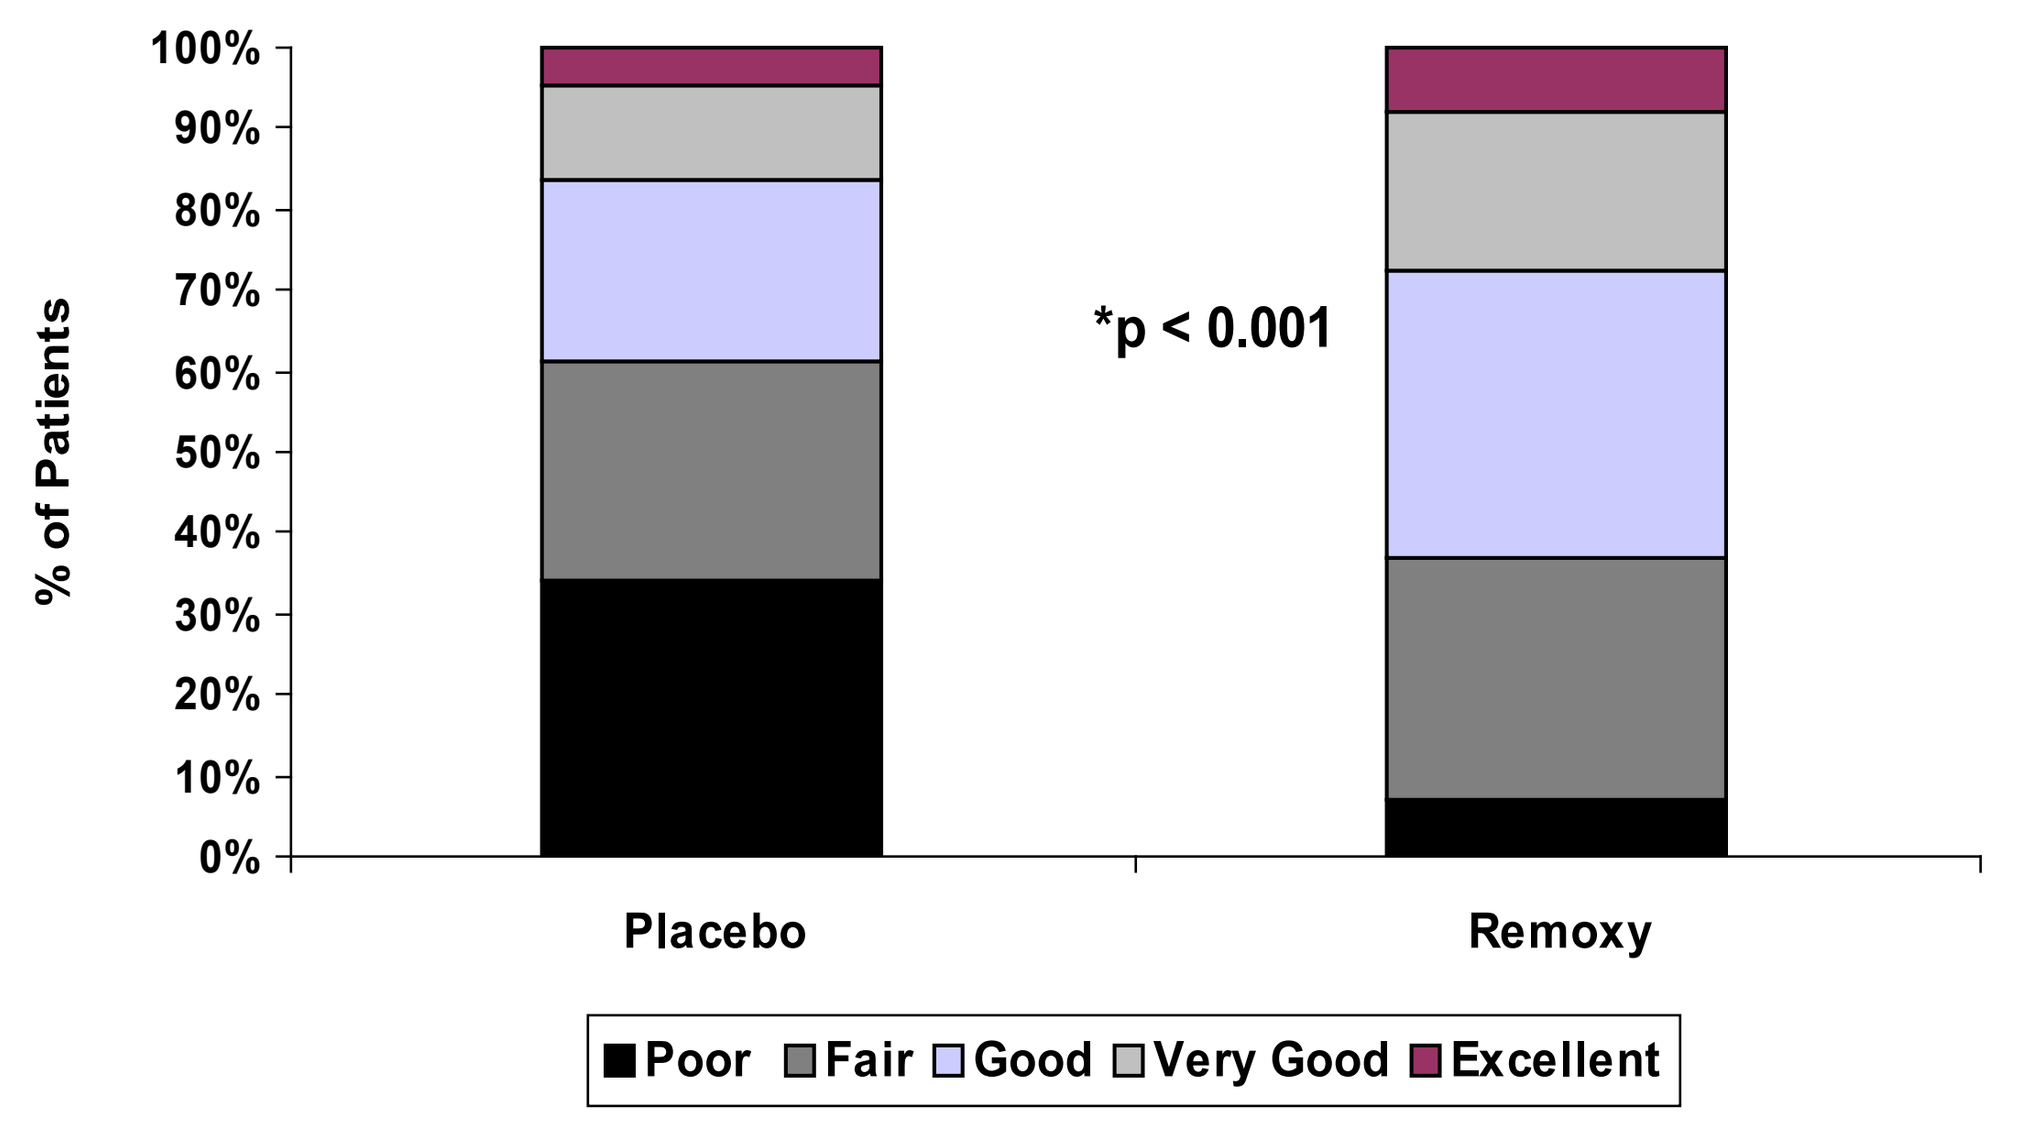

% of Patients
*p < 0.001
Placebo
Remoxy
Poor
Fair
Good
Very Good
Excellent

Functional Assessments

Patients taking Remoxy vs. placebo had superior functional improvement indicated by the WOMAC Osteoarthritis Index pain intensity subscale (p=0.015), stiffness subscale (p=0.040) and total score (p=0.045; Fig. 6). The SF-12 physical component scale indicated greater improvement in quality of life for the Remoxy group compared to the placebo group (p<0.001; Fig. 5). There were no significant differences between treatment groups for the mental component scale of the SF-12.

Fig. 6 **WOMAC Subscales and Total Score**

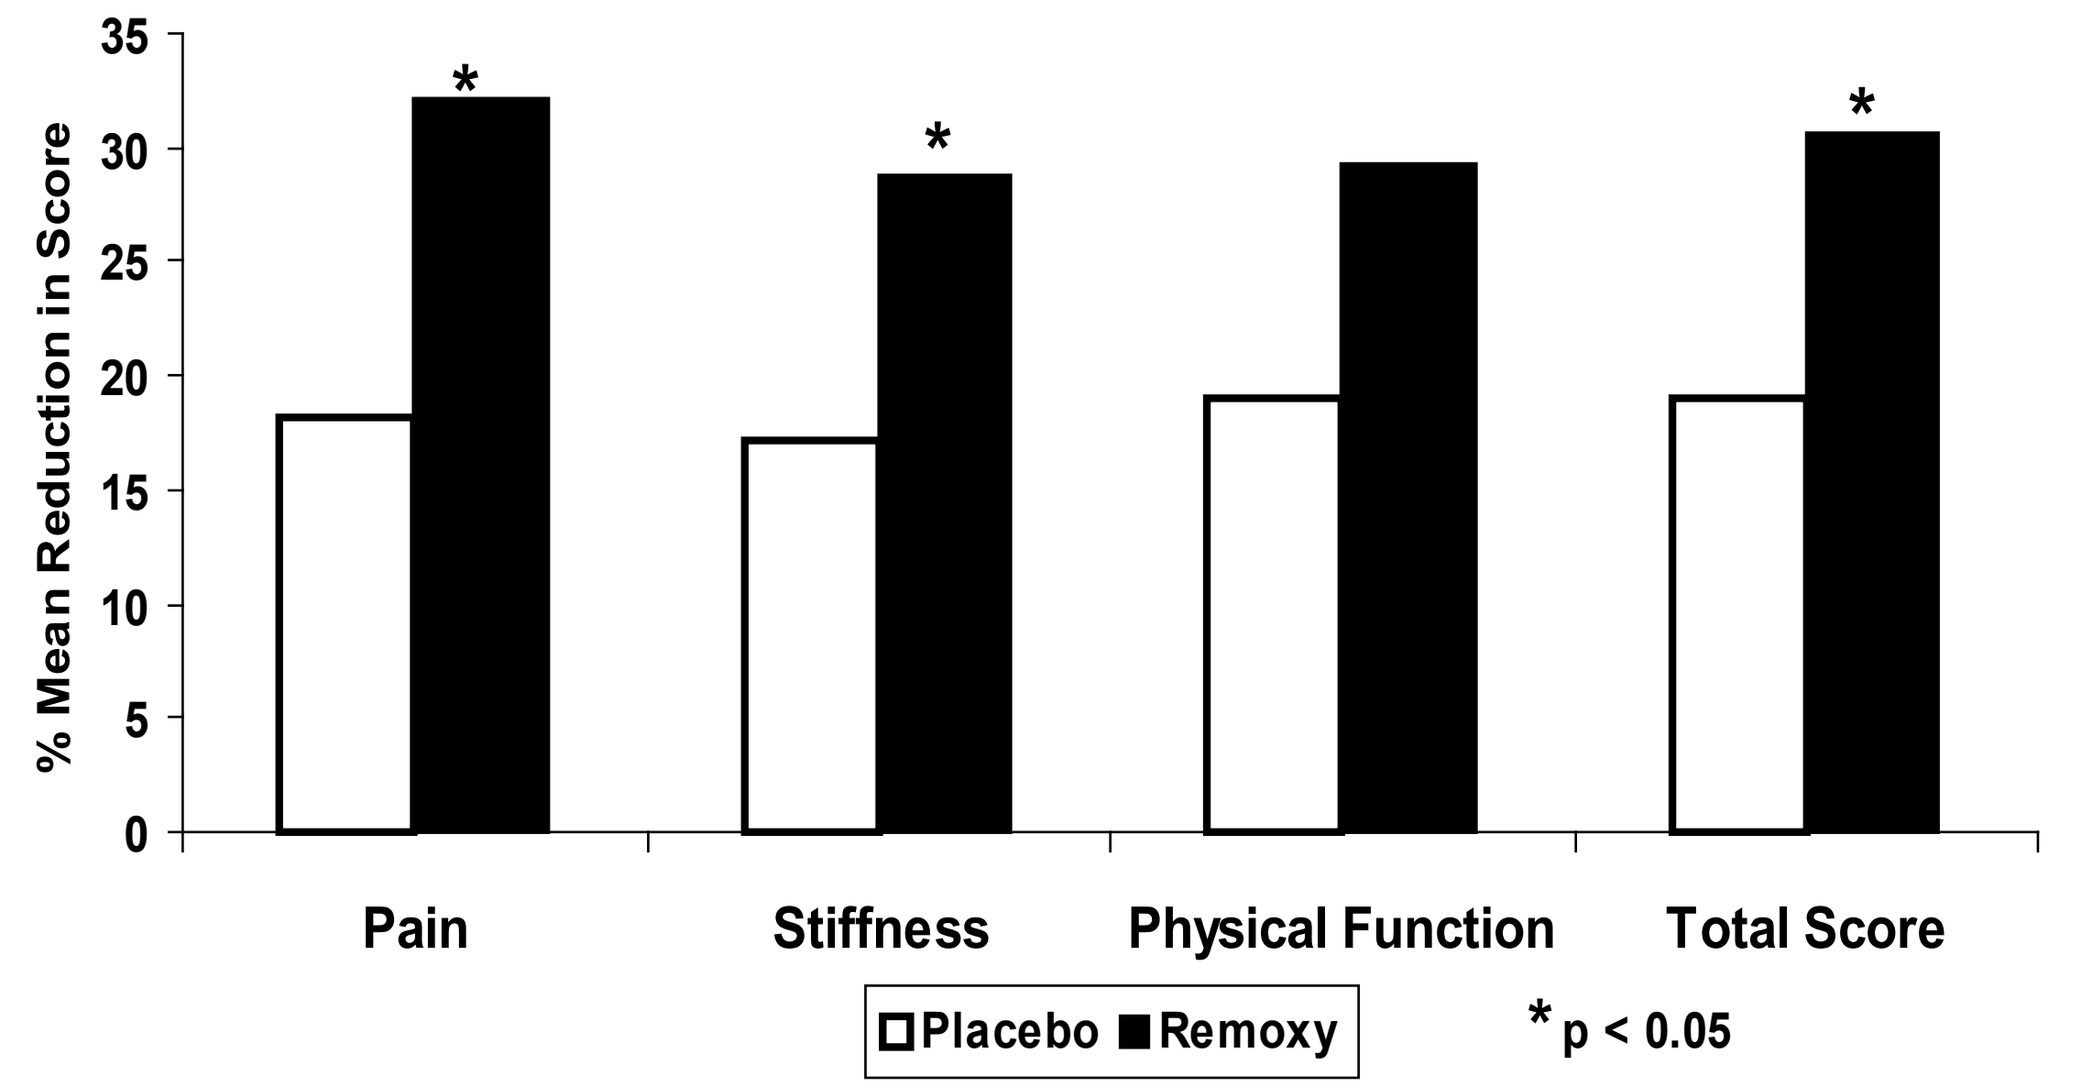

% Mean Reduction in Score
Pain
Stiffness
Physical Function
Total Score
Placebo
Remoxy
* p < 0.05

Adverse Events

No drug-related serious adverse events were reported. As expected, adverse events reported in the Remoxy treatment group were typical opioid-related side effects such as nausea/vomiting, pruritis, drowsiness, constipation, and dizziness.

DISCUSSION

The present double-blind, randomized Phase III clinical trial data show that twice-daily administration of Remoxy provides effective analgesia compared to placebo in patients with chronic moderate-to-severe pain due to osteoarthritis. Significant differences were obtained on the primary endpoint, percent reduction in Pain Intensity by the end of treatment, as well as on secondary endpoints of Quality of Analgesia, and WOMAC pain and stiffness subscales and WOMAC total score. Previous pharmacokinetic studies have shown that unlike currently marketed controlled-release formulations of oxycodone, Remoxy does not yield any substantial portion of its 12-hr dose if abused by crushing and dissolving in water, alcohol or other common beverages.

CONCLUSION

This clinical trial demonstrates that twice-daily administration of Remoxy provides effective, long-acting analgesia and improved quality of life in chronic pain patients with osteoarthritis. Remoxy provides additional safety compared to currently marketed controlled-release oxycodone products since Remoxy's proprietary long-acting formulation resists abuse by crushing or dissolving in alcohol or other beverages.